NEUROIMAGING OF M4 MUSCARINIC ACETYLCHOLINE RECEPTORS USING [^{11}C]MK-6884 IN RHESUS MACAQUES

Vasily Belov[1], Nicolas J. Guehl[1], Sridhar Duvvuri[2], Philip Iredale[2], Sung-Hyun Moon[1], Maeva Dhaynaut[1], Peter A. Rice[1], Daniel L. Yokell[1], John Renger[2], Georges El Fakhri[1], Marc D. Normandin[1]

[1]Gordon Center for Medical Imaging, Massachusetts General Hospital, Harvard Medical School, Boston, MA; [2]Cerevel Therapeutics, LLC, Cambridge, MA

RATIONALE

- M4 subtype muscarinic acetylcholine receptors (mAChR) are implicated in the regulation of striatal acetylcholine and dopamine equilibrium, imbalance of which leads to neurodegenerative and psychiatric disorders. It is hypothesized that M4 mAChR stimulation alleviates psychotic events by reducing striatal hyperdopaminergia.
- Investigation of individual muscarinic receptors is challenging due to their high degree of homology at the acetylcholine binding site. Recent development of [^{11}C]MK-6884, a PET radioligand selectively binding to the allosteric site of M4 mAChR, opens attractive opportunities for quantitative assessment of target engagement of M4 positive allosteric modulator (PAM) drugs by PET, provided imaging performance of [^{11}C]MK-6884 is sufficiently characterized.
- CVL-231 is a novel, brain-penetrant, highly selective PAM of M4 mAChR being developed by Cerevel for the treatment of neurobehavioral components, such as delusions, hallucinations, and conceptual disorganization, associated with schizophrenia and other neurodegenerative diseases. Translation of the drug to early clinical development necessitated evaluation of the relationship between plasma exposure and M4 mAChR occupancy by the drug in the brain regions of non-human primates (NHP).

OBJECTIVES

1. **Characterize PET imaging and quantitative performance of [^{11}C]MK-6884 using arterial input function-based and reference tissue-based kinetic modelling methods.**
2. **Determine the receptor occupancy (RO) of CVL-231 in relationship to the dose and plasma exposure of the drug.**

METHODS

Study design

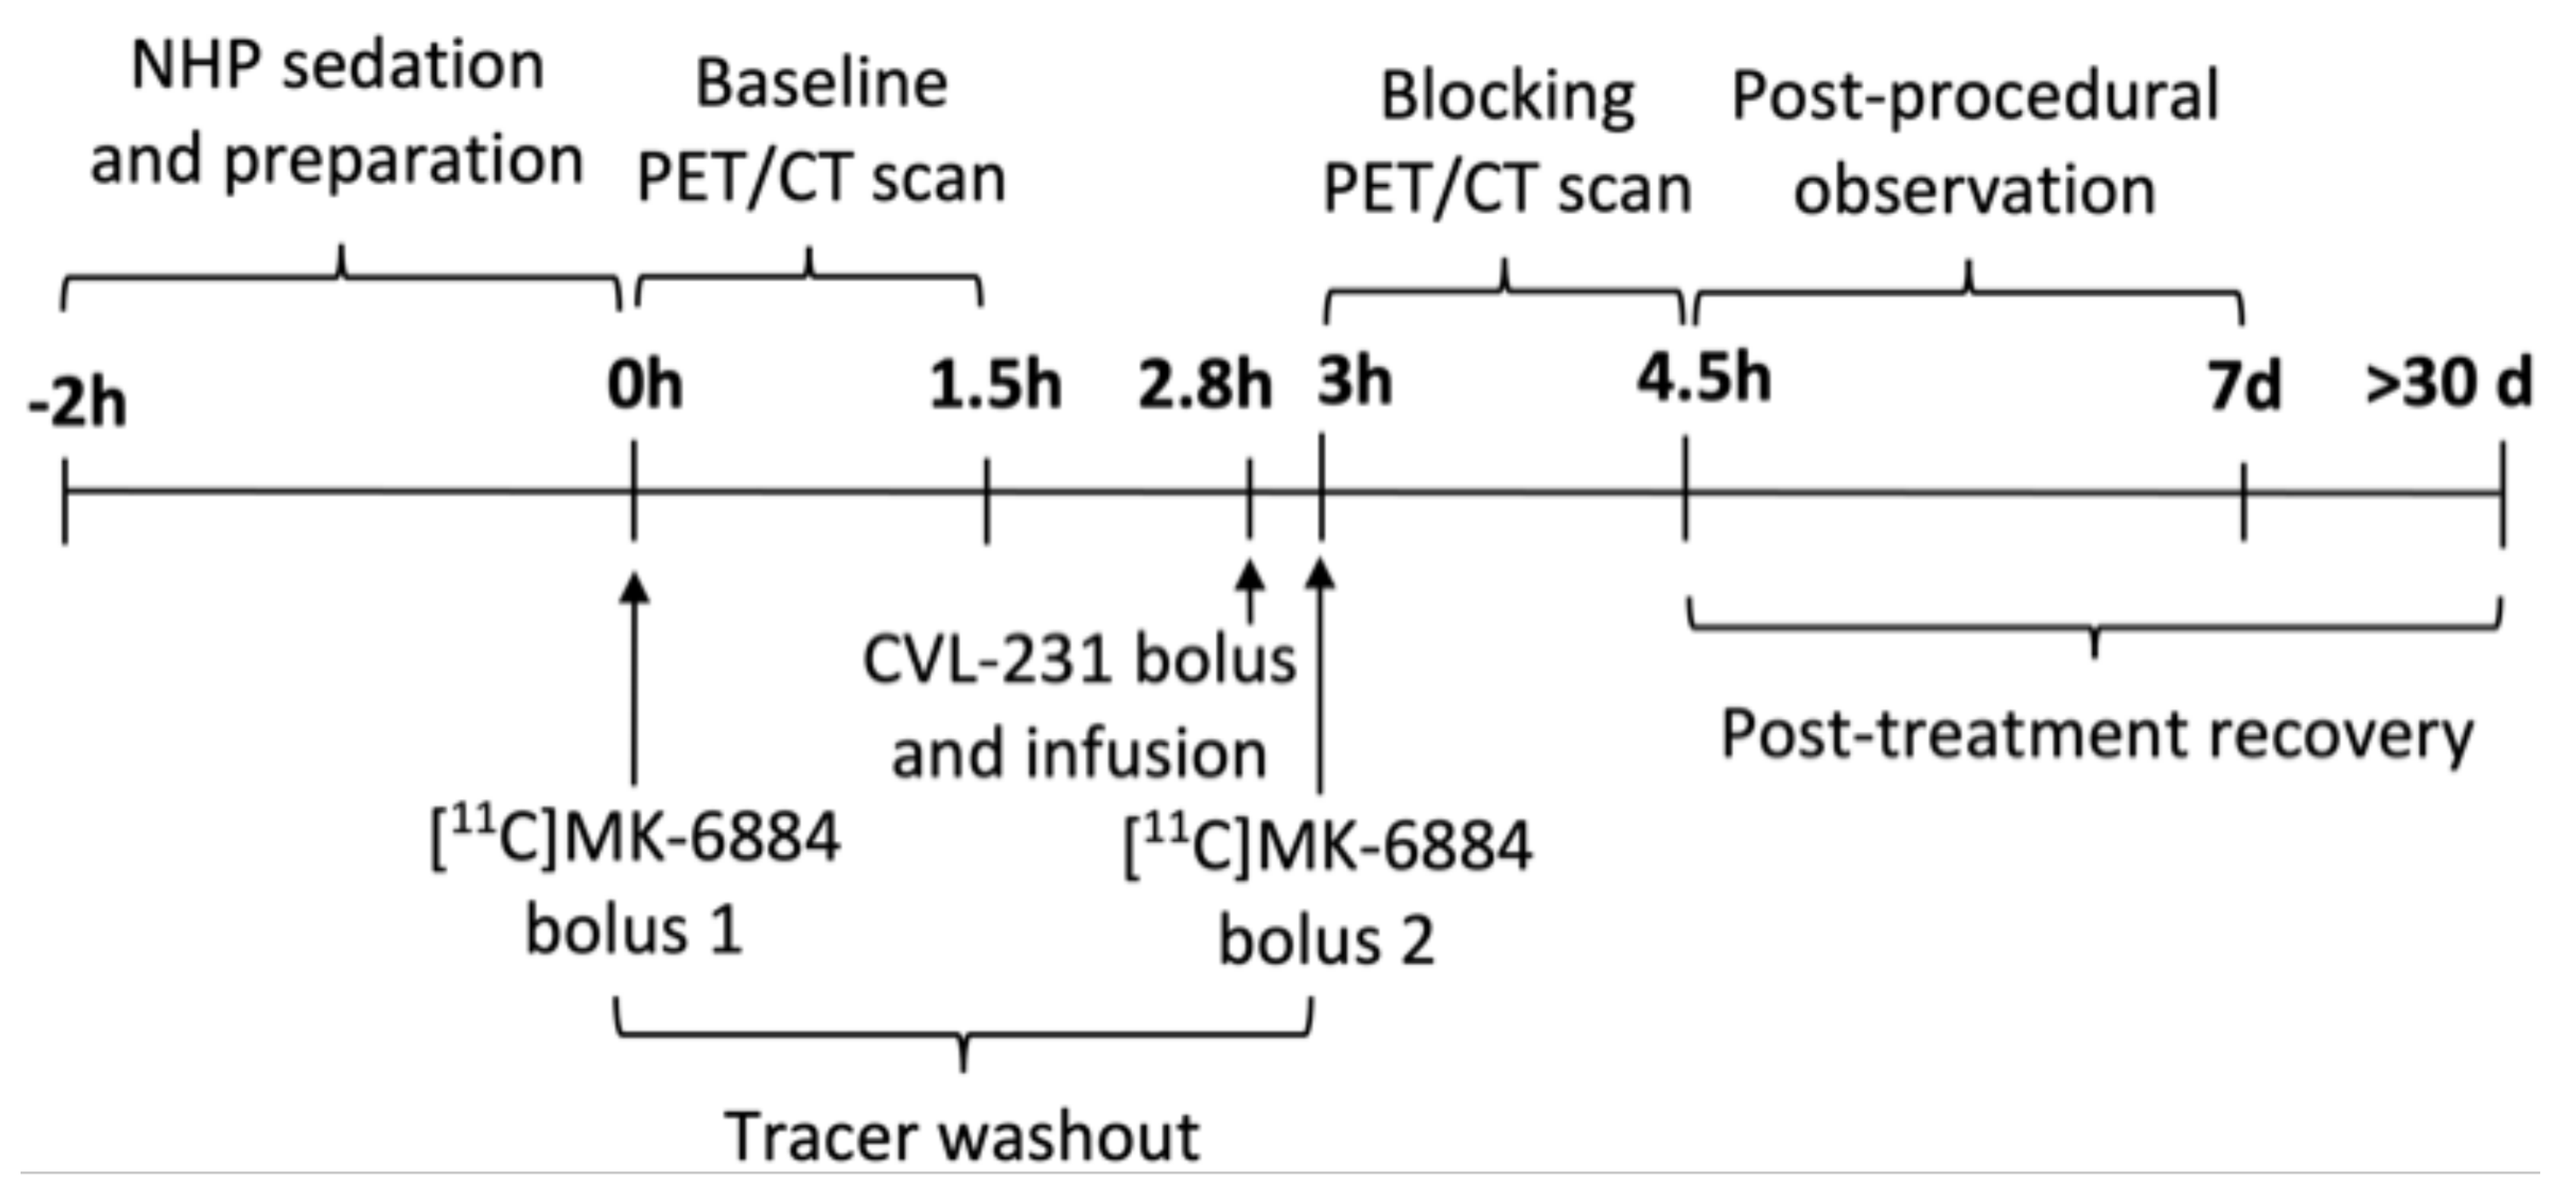
NHP sedation and preparation
Baseline PET/CT scan
Blocking PET/CT scan
Post-procedural observation
-2h
0h
1.5h
2.8h
3h
4.5h
7d
>30 d
CVL-231 bolus and infusion
[11C]MK-6884 bolus 1
[11C]MK-6884 bolus 2
Post-treatment recovery
Tracer washout

Radiosynthesis

[^{11}C]MK-6884 was synthesized with high molar activity (187.2±68.7 GBq/µmol) and radiochemical purity (100%) by reaction of the precursor, L-005388167, with [^{11}C]CH_3I followed by serial HPLC purification, washing in Waters Sep-Pak C-18 Light SPE cartridge and final formulation in 0.9% sodium chloride for injection, USP.

Animals

2 rhesus macaques (*m. mulatta*): Monkey 1 (M1), 9 y.o., BW: 12.9 kg [12.2;13.4]; Monkey 2 (M2), 13 y.o., BW: 15.1 kg. M1 underwent 3 paired baseline-blocking studies and M2 4 using various CVL-231 doses.

[^{11}C]MK-6884 and CVL-231 dose administration

- [^{11}C]MK-6884 was administered via the lateral saphenous vein over a 3 min infusion followed by a 3-min infusion of a saline flush using syringe pumps (Medfusion 3500). The injected radioactive dose at the time of injection was 5.11 ± 0.40 mCi ([4.61;5.87], n = 12). Specific activity (SA) at the end of synthesis was 5.06 ± 1.86 Ci/µmol ([2.19;7.58], n = 12) corresponding to the injected masses of 1.33 ± 1.02 µg ([0.50;4.30], n = 12).
- CVL-231 was injected at 0.25, 0.5, and 1.7 mg/kg in M1 and 0.5, 1.0, 1.7, and 3.37 mg/kg in M2. All administrations were performed in two steps: 2-3 min bolus of loading dose (~48%) 10 min prior scan start + constant infusion of maintenance dose (~52%) until the end of scan.

Imaging and blood data collection

PET/CT: Discovery MI (GE Healthcare) scanner, 90-min dynamic acquisition, followed by reconstruction by a 3D TOF OSEM method, which includes 3 iterations and 34 subsets with point-spread function modeling and corrections for scatter and attenuation photons, deadtime events, and random coincidences.
MRI: T1-weighted magnetization-prepared rapid gradient-echo (MPRAGE) images acquired on 3T Biograph mMR (Siemens Medical Systems) for each monkey once.
Arterial sampling: 1-3 ml serial arterial samples were collected during each PET scan for the measurement of time course of radioactivity concentration in whole blood and plasma and for radiometabolite analysis using an automated column-switching radioHPLC system. Also, 3 ml samples were collected during each blocking scan at 60 min post [^{11}C]MK-6884 injection and used for measurement of CVL-231 concentration in plasma.

Data processing and analysis

- Both metabolite-corrected plasma input function-based (1T2k, 2T4k, Logan plot, MA1) and reference region-based (SRTM, SRTM2, MRTM2 and Logan DVR) modelling methods were applied to calculate total distribution volume (V_T) and binding potential (BP_{ND}) in various cerebral regions (caudate nucleus, cerebellar grey matter, entire cortex, hippocampus, putamen, central white matter).
- RO was quantified using (1) Lassen plot: $V_T^{baseline} - V_T^{blocking} = RO * (V_T^{baseline} - V_{ND})$; and (2) BP_{ND} estimates: $\%RO = 100\% \times (1 - BP_{ND}^{blocking}/BP_{ND}^{baseline})$

RESULTS

Blood data

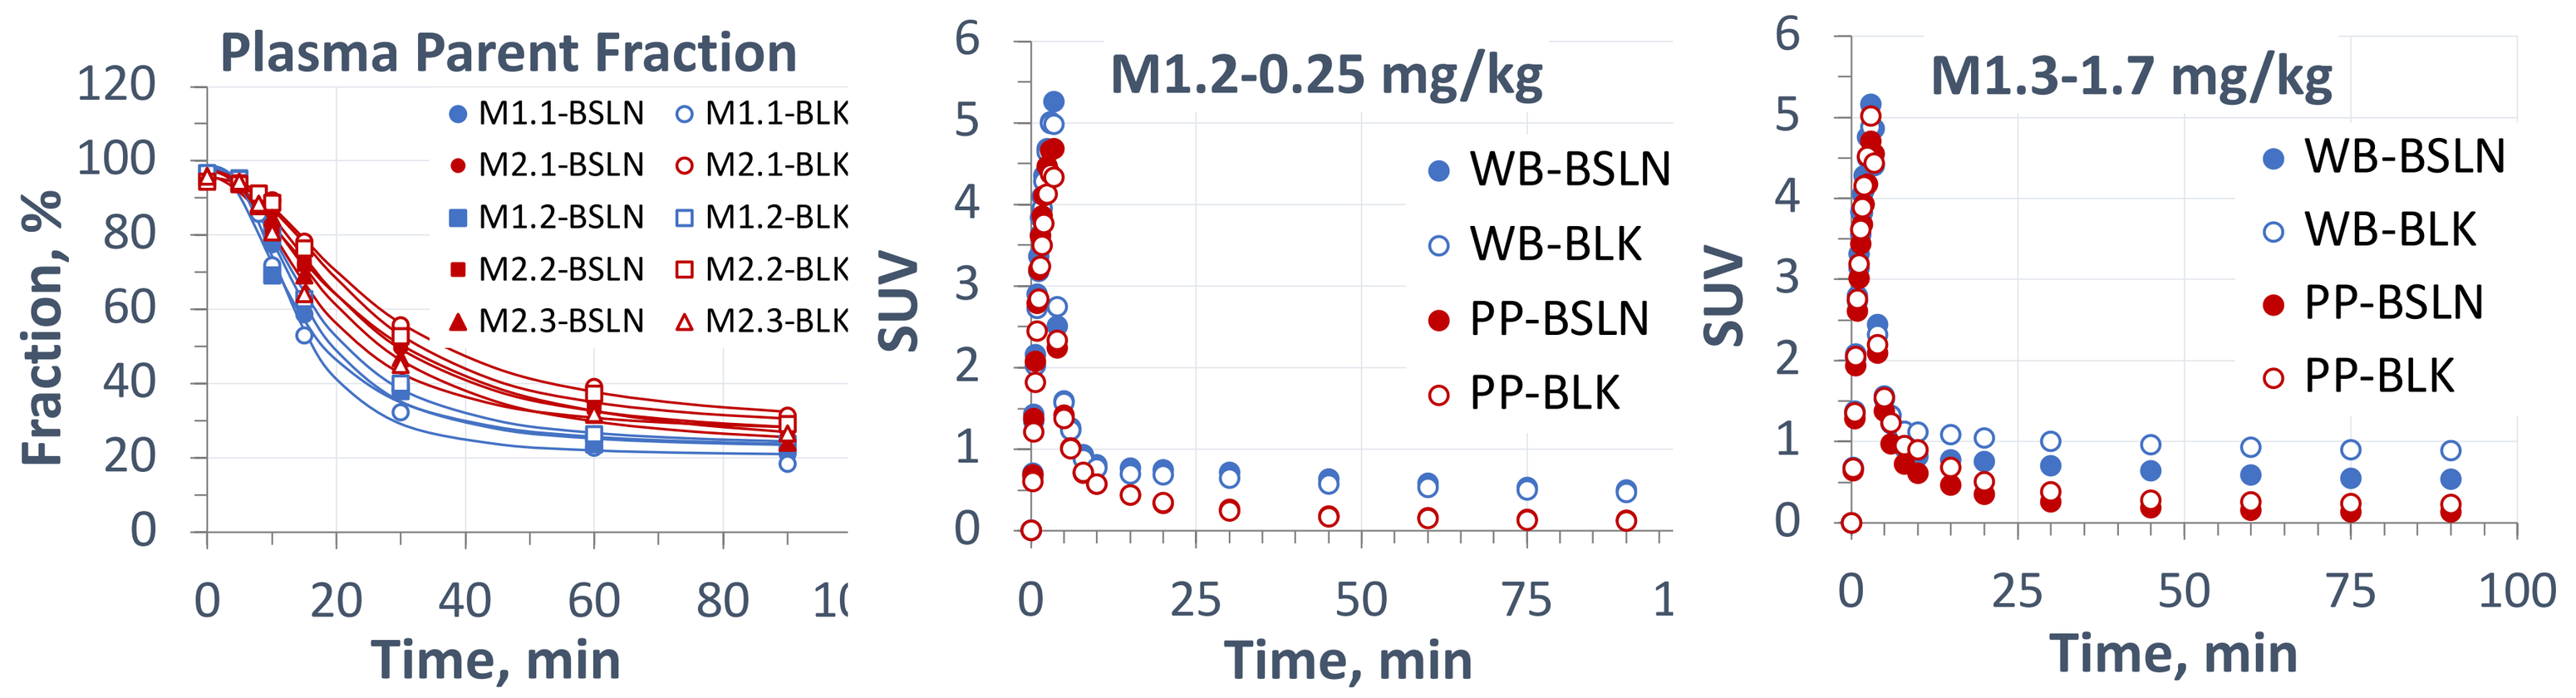
Plasma Parent Fraction
M1.1-BSLN
M1.1-BLK
M2.1-BSLN
M2.1-BLK
M1.2-BSLN
M1.2-BLK
M2.2-BSLN
M2.2-BLK
M2.3-BSLN
M2.3-BLK
Fraction, %
Time, min
M1.2-0.25 mg/kg
M1.3-1.7 mg/kg
WB-BSLN
WB-BLK
PP-BSLN
PP-BLK
SUV

Left to right: Time course of the unmetabolized fraction in plasma. Representative time courses of the total radioactivity concentrations in whole blood and of unmetabolized tracer in plasma (input function) during respective baseline and blocking scans for the lowest and highest doses of CVL-231.

Blood-based analysis of the regional uptake

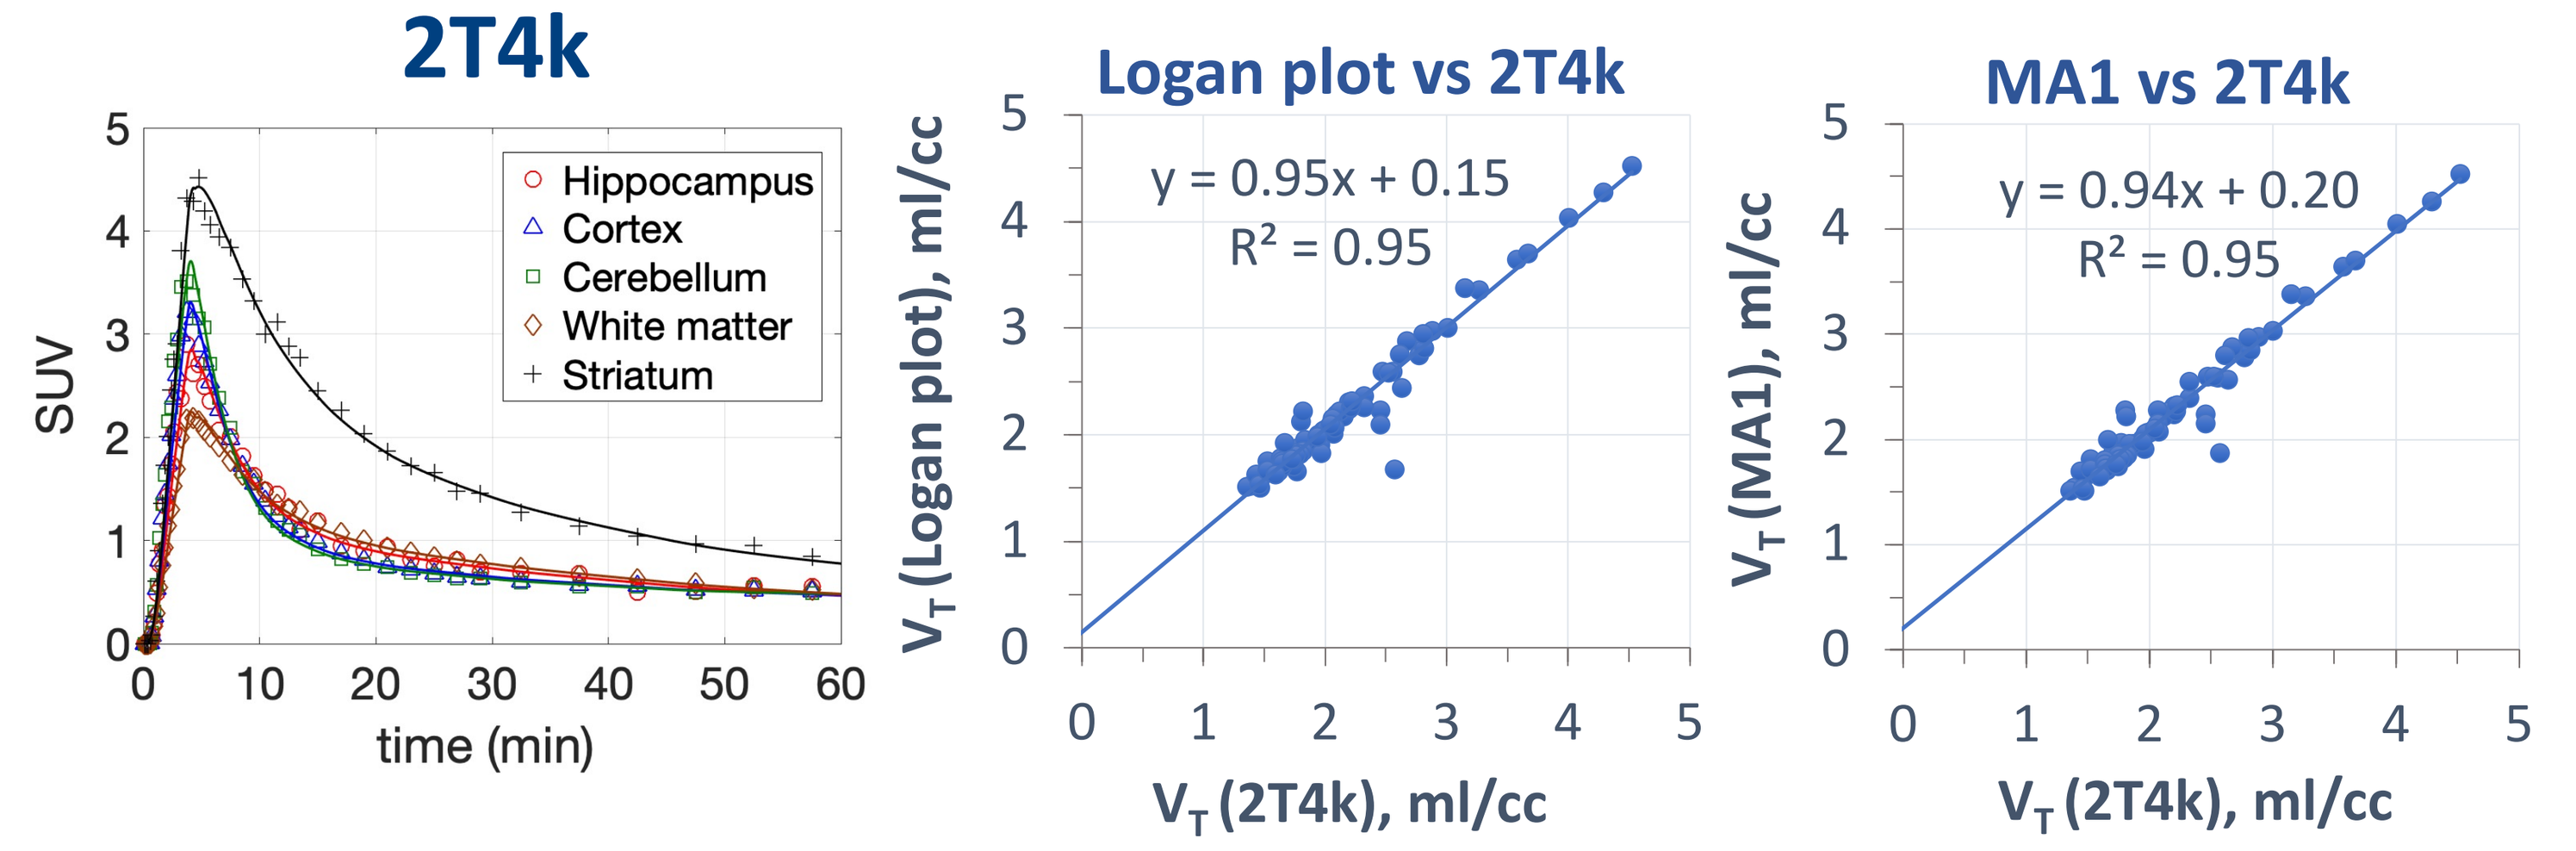
2T4k
Hippocampus
Cortex
Cerebellum
White matter
Striatum
SUV
time (min)
Logan plot vs 2T4k
y = 0.95x + 0.15
R² = 0.95
VT (Logan plot), ml/cc
VT (2T4k), ml/cc
MA1 vs 2T4k
y = 0.94x + 0.20
R² = 0.95
VT (MA1), ml/cc

Left to right: representative SUV TACs with 2T4k model fits. Agreement of the V_T estimates by different blood-based methods.

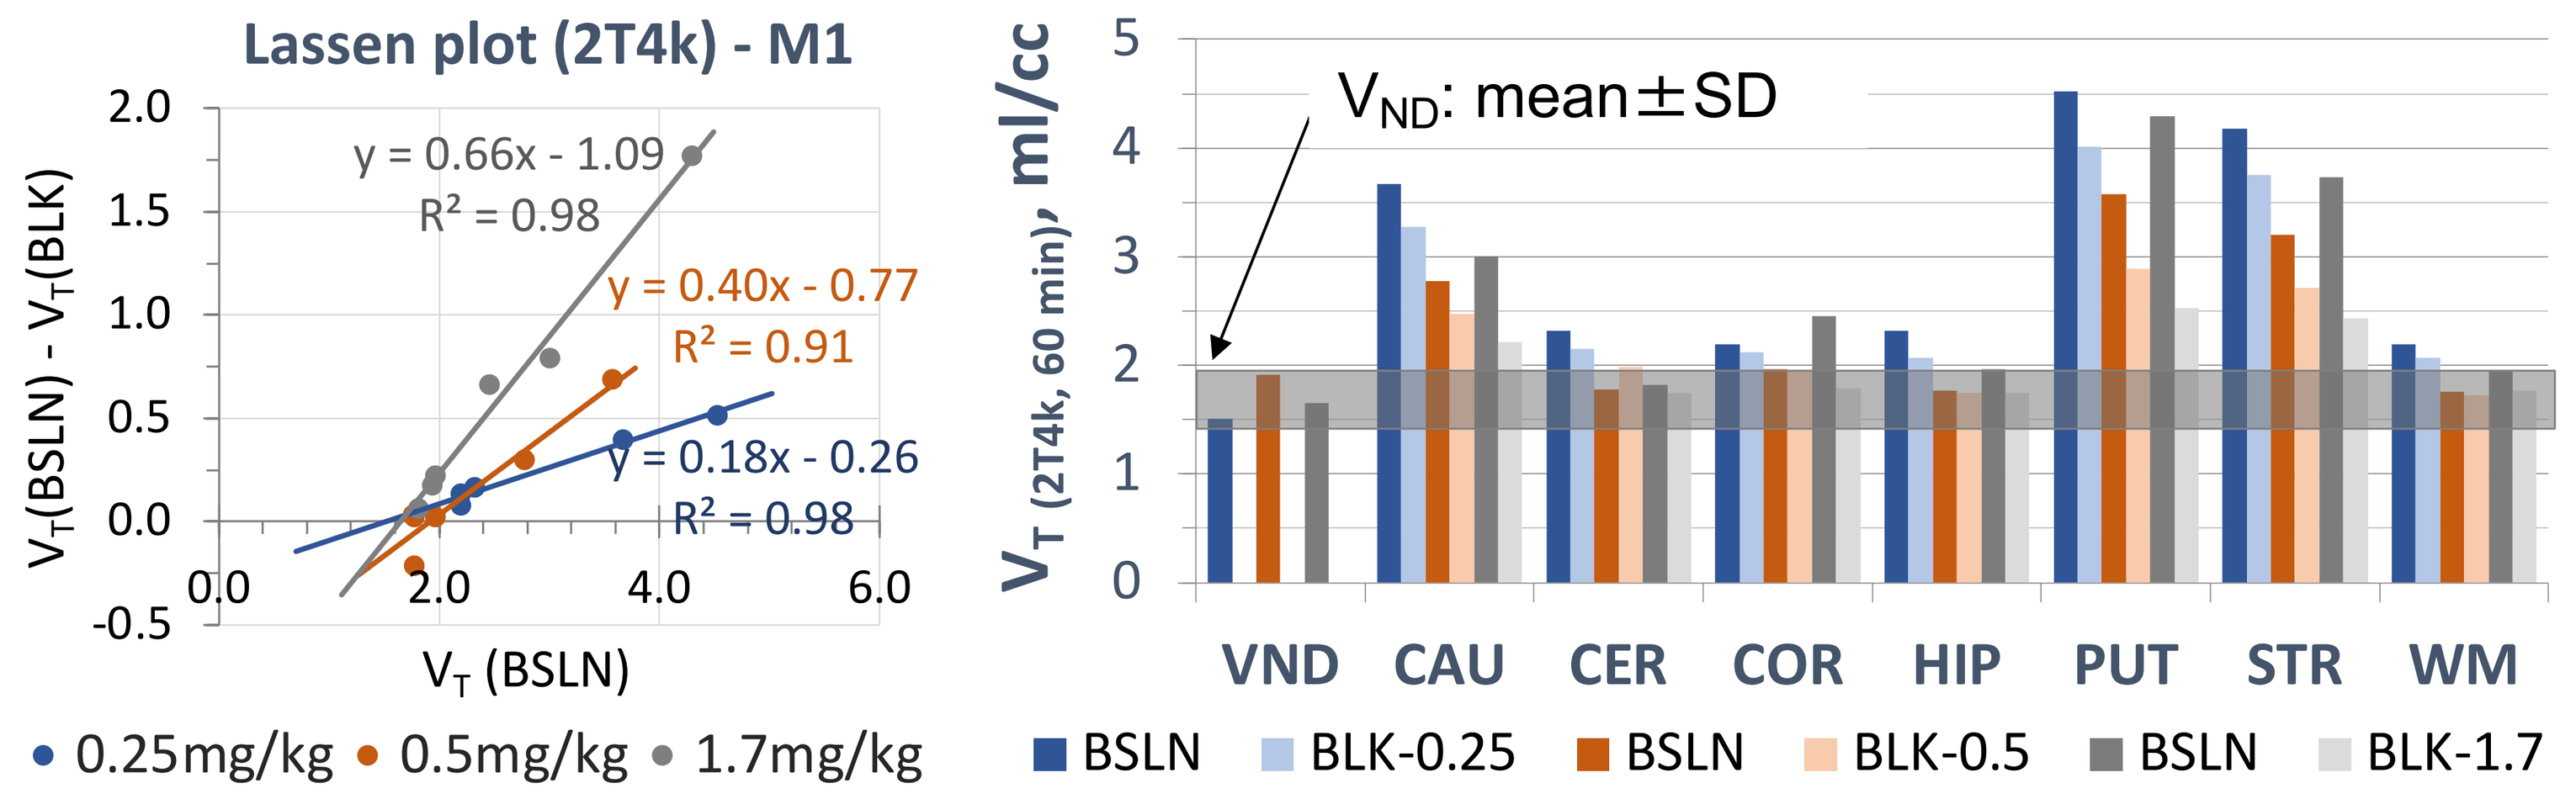
Lassen plot (2T4k) - M1
y = 0.66x - 1.09
R² = 0.98
y = 0.40x - 0.77
R² = 0.91
y = 0.18x - 0.26
R² = 0.98
VT(BSLN) - VT(BLK)
VT (BSLN)
0.25mg/kg
0.5mg/kg
1.7mg/kg
VND: mean±SD
VT (2T4k, 60 min), ml/cc
VND
CAU
CER
COR
HIP
PUT
STR
WM
BSLN
BLK-0.25
BLK-0.5
BLK-1.7

Left: representative Lassen plot analysis for monkey 1. Right: regional V_T in monkey 1 obtained by 2T4k. The shaded area represents mean±SD of non-displaceable volume of distribution (V_{ND}) obtained by Lassen plot analysis.

Reference region-based analysis

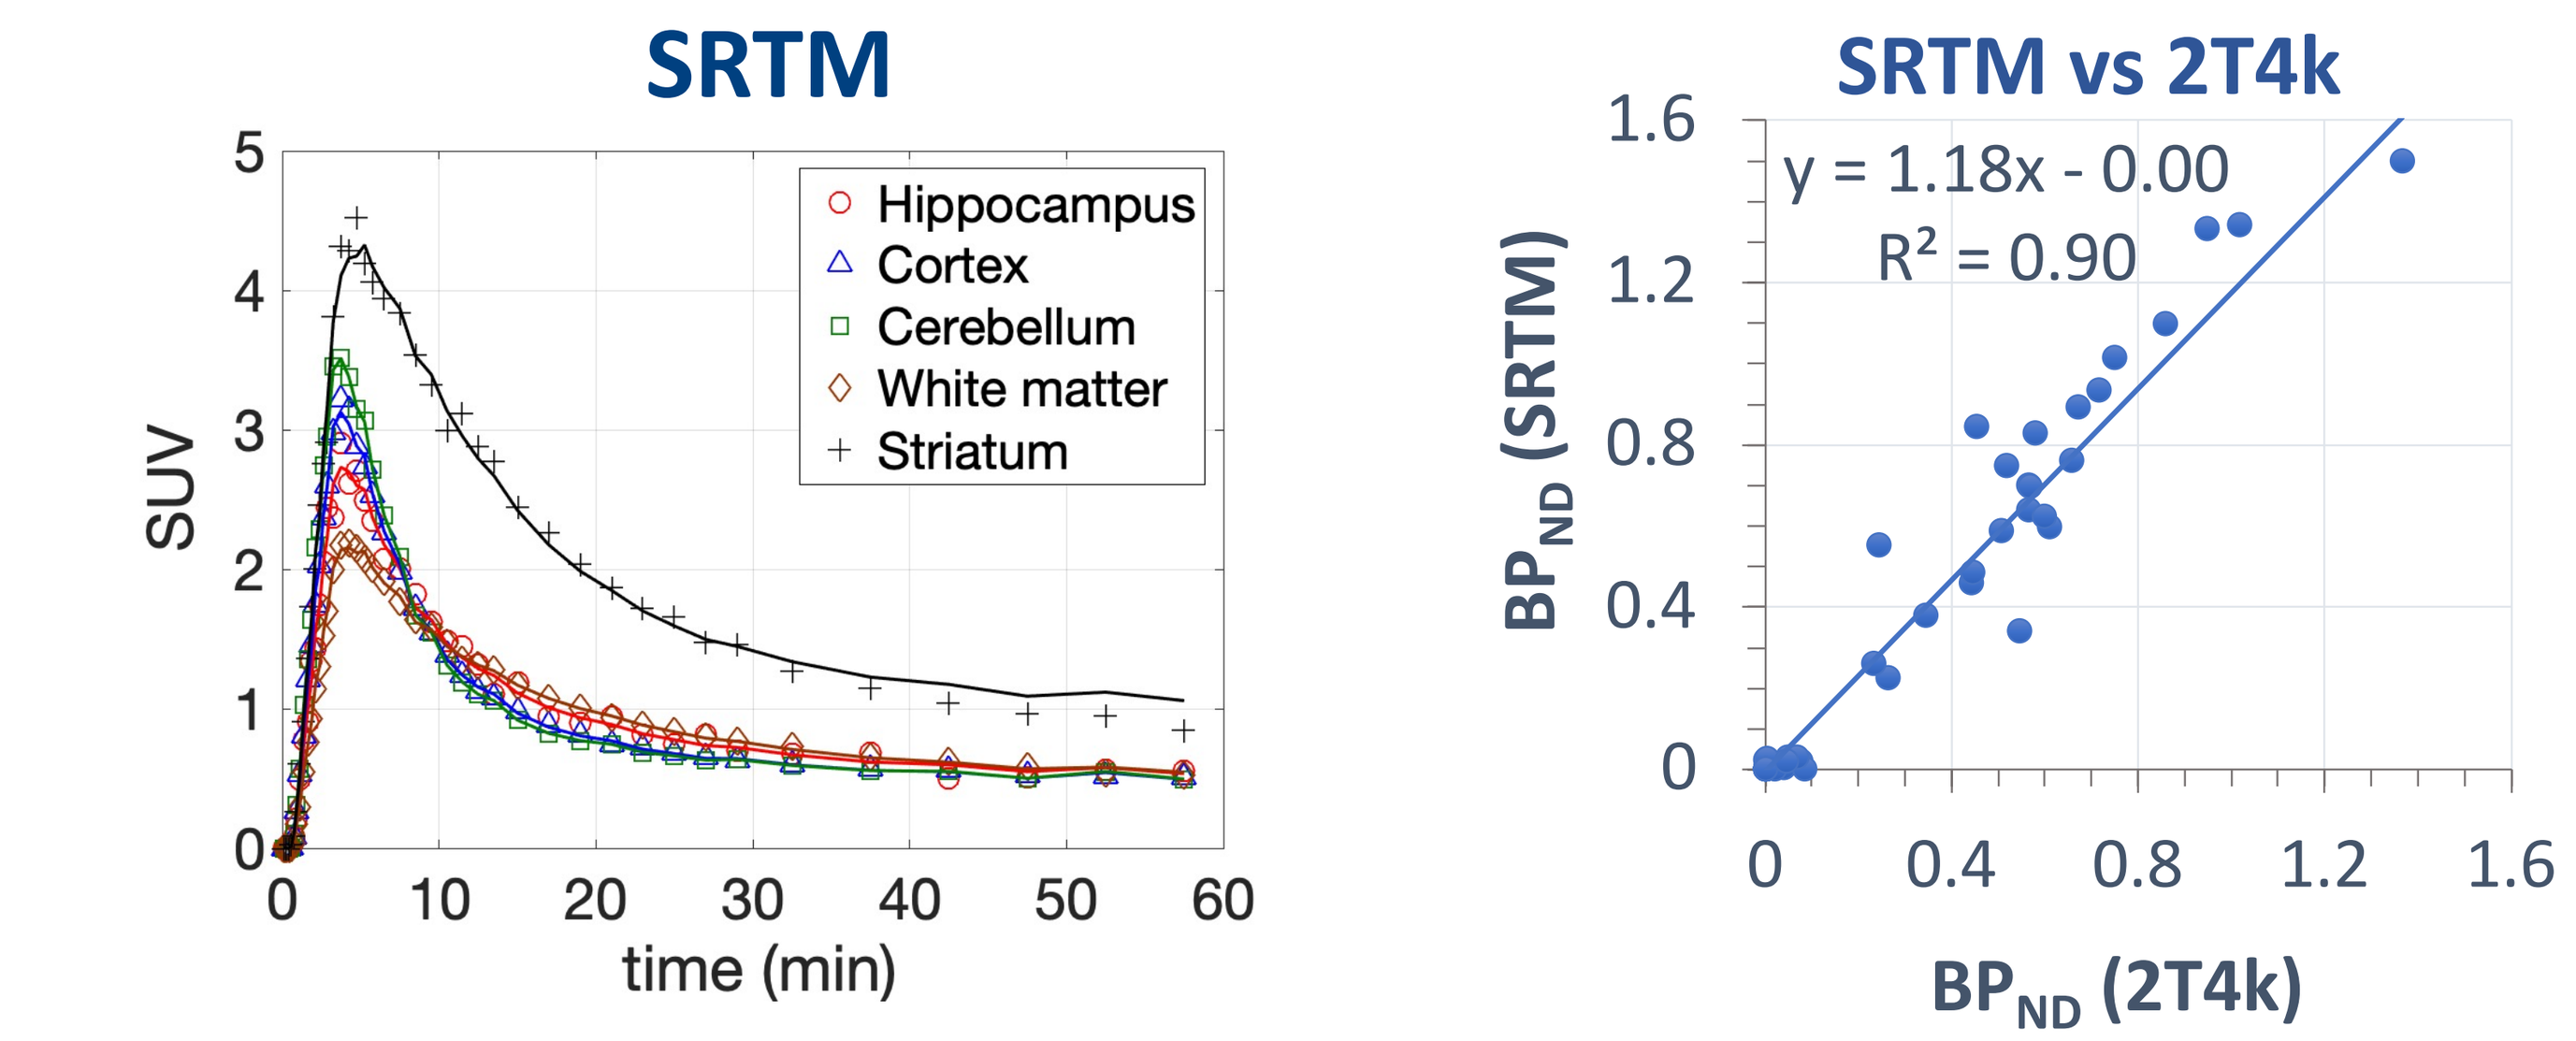
SRTM
Hippocampus
Cortex
Cerebellum
White matter
Striatum
SUV
time (min)
SRTM vs 2T4k
y = 1.18x - 0.00
R² = 0.90
BPND (SRTM)
BPND (2T4k)

Striatal receptor occupancy by CVL-231

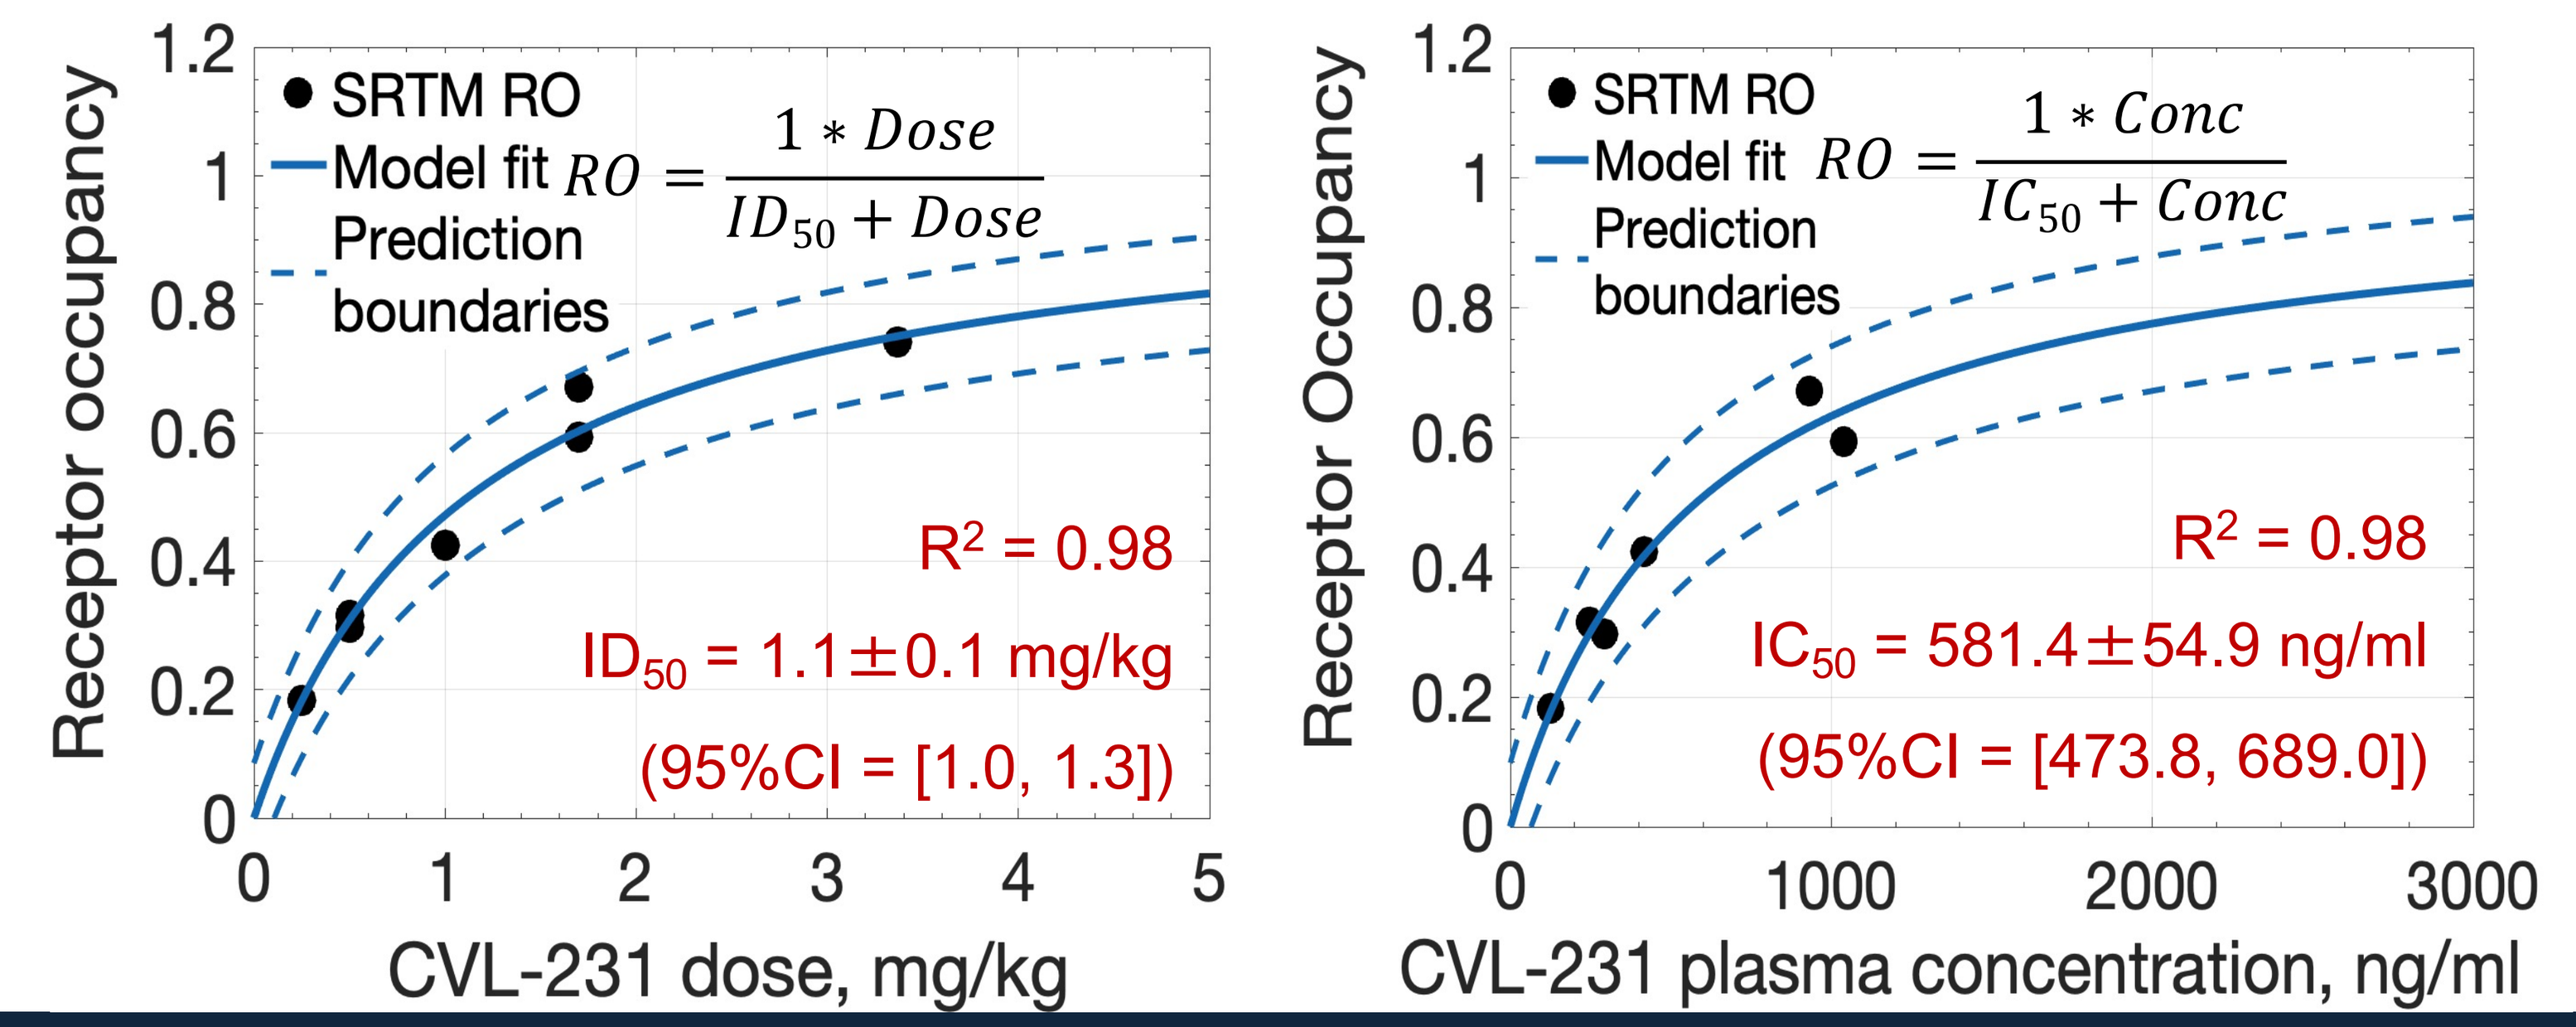
SRTM RO
Model fit
Prediction boundaries
$RO = \frac{1 * Dose}{ID_{50} + Dose}$
R² = 0.98
ID50 = 1.1±0.1 mg/kg
(95%CI = [1.0, 1.3])
Receptor occupancy
CVL-231 dose, mg/kg
$RO = \frac{1 * Conc}{IC_{50} + Conc}$
R² = 0.98
IC50 = 581.4±54.9 ng/ml
(95%CI = [473.8, 689.0])
Receptor Occupancy
CVL-231 plasma concentration, ng/ml

Parametric images

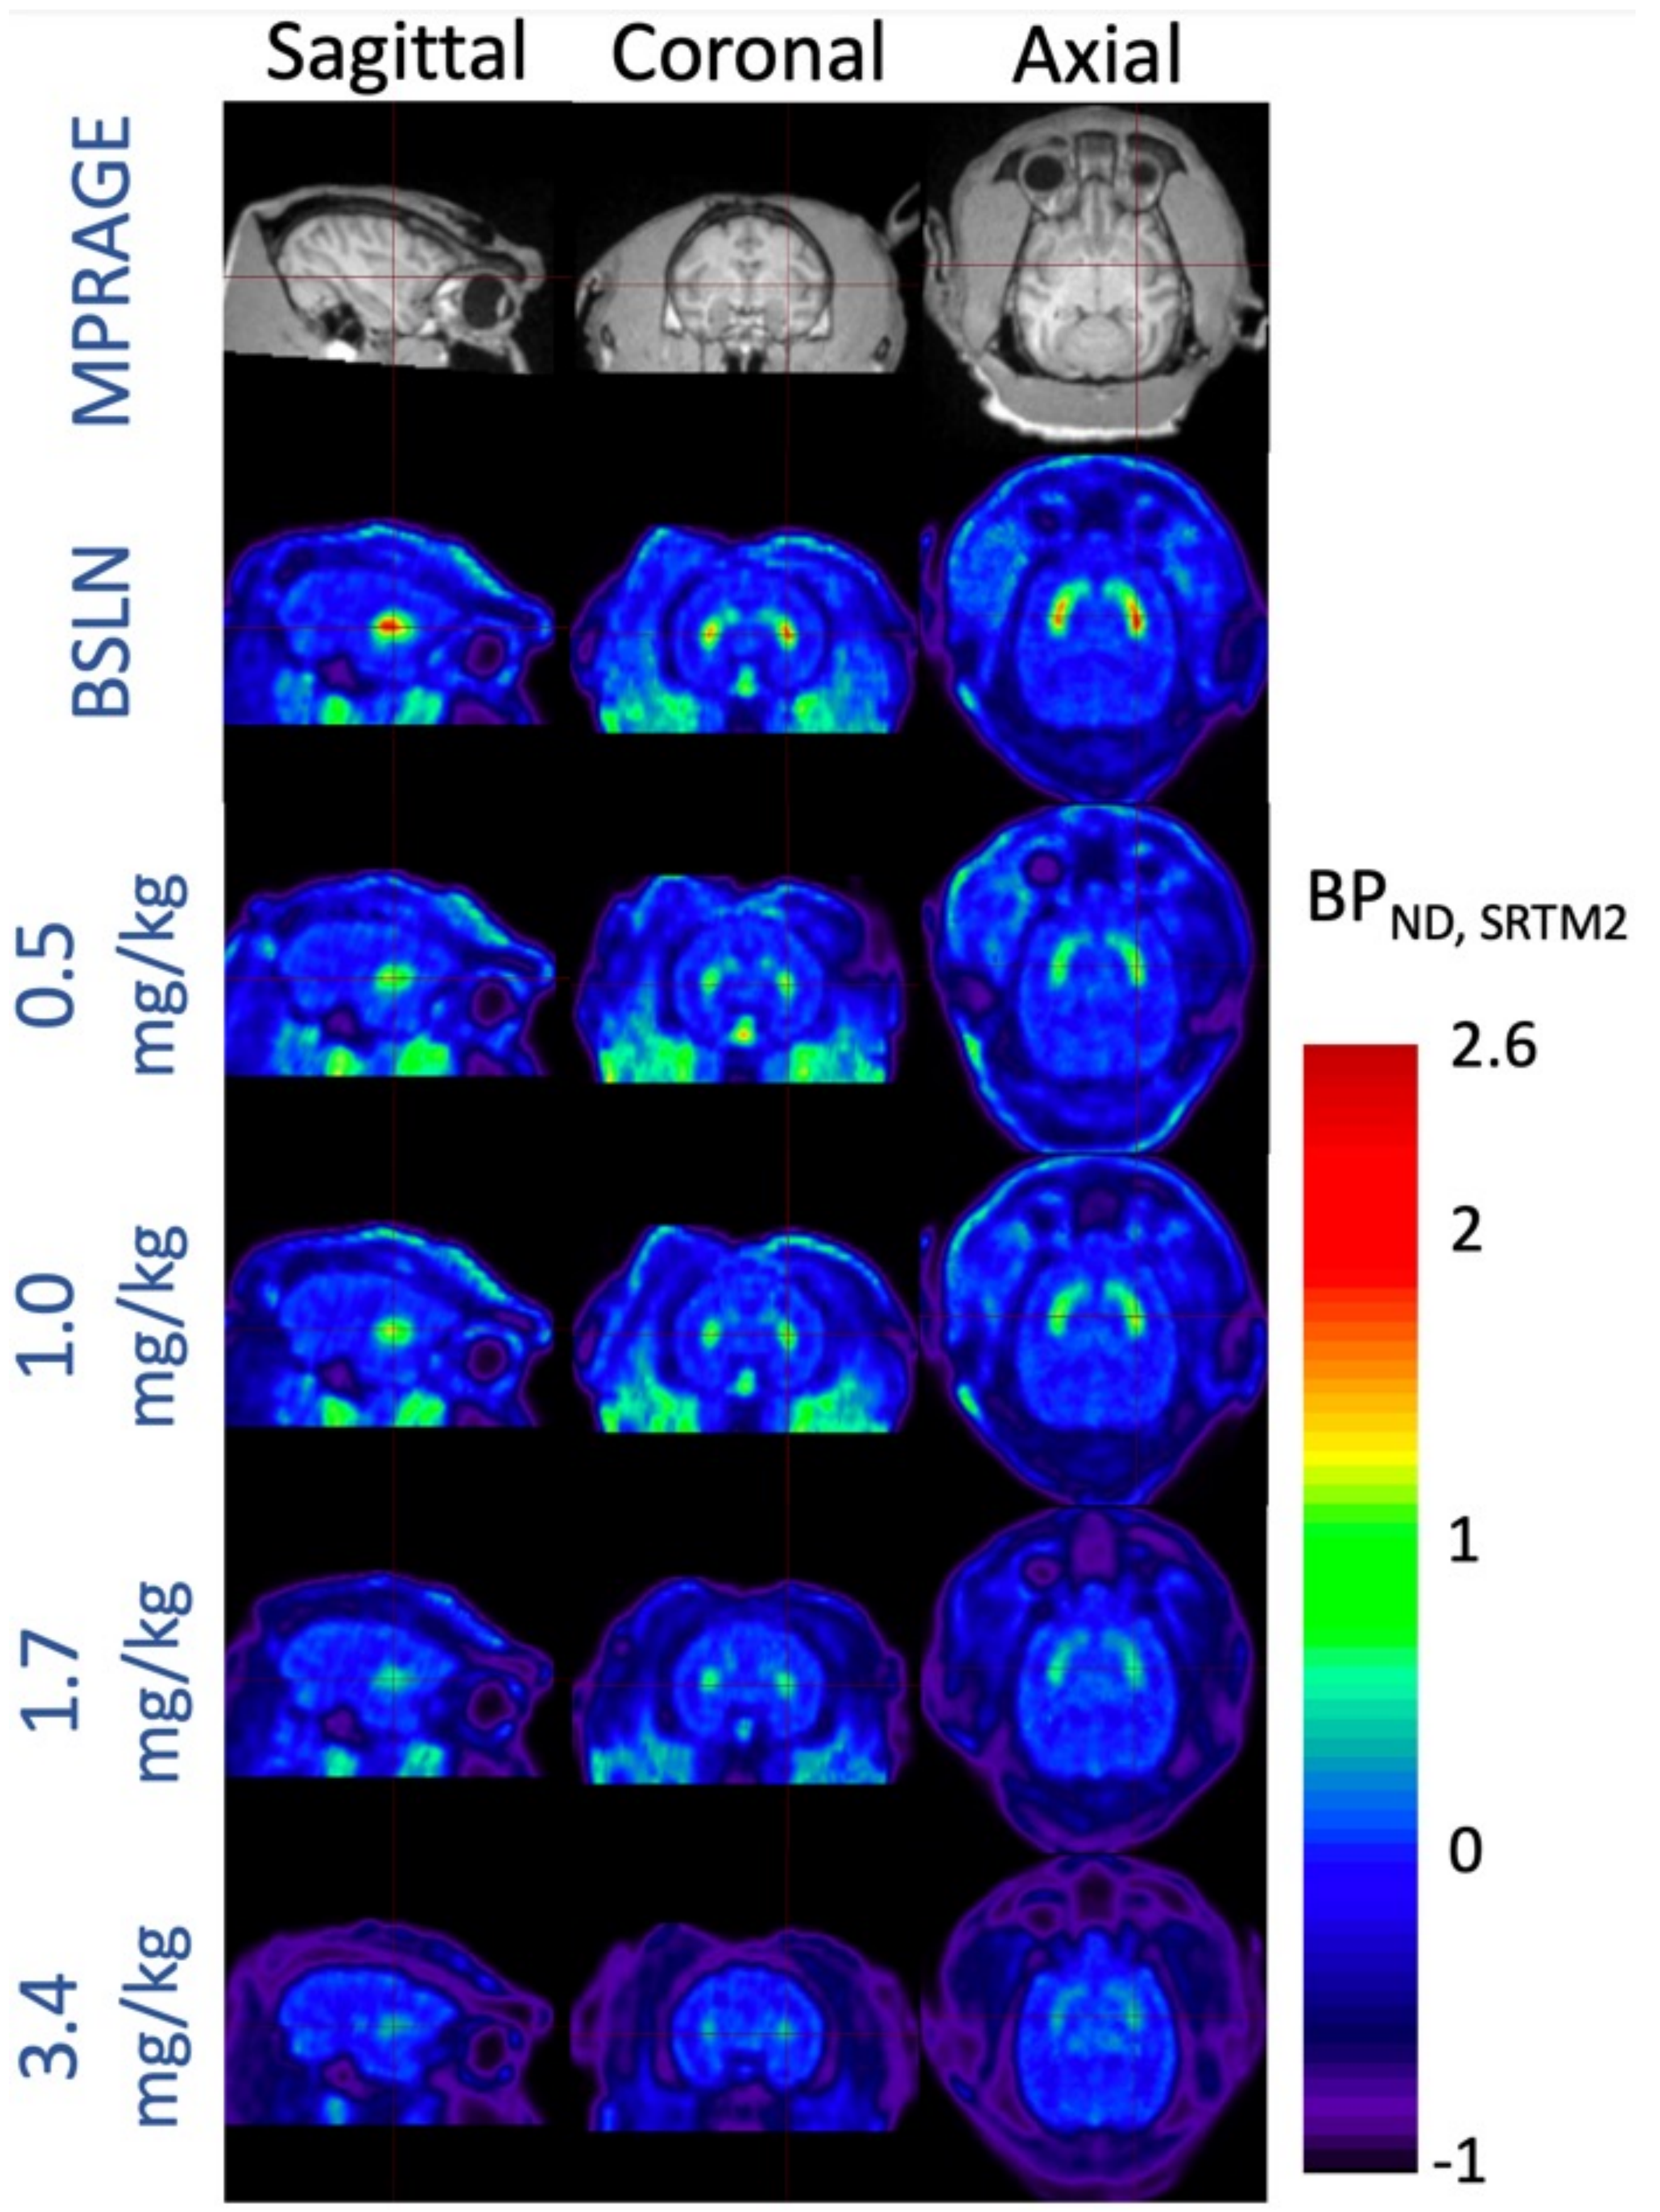
Sagittal
Coronal
Axial
MPRAGE
BSLN
0.5 mg/kg
1.0 mg/kg
1.7 mg/kg
3.4 mg/kg
BPND, SRTM2
2.6
2
1
0
-1

Representative PET parametric maps of BP_{ND} obtained by SRTM2 method using fixed k_2' parameter for one baseline scan and each CV-231 dose injected in monkey 2. All maps are co-registered with the MRI MEMPRAGE image using affine image transformations to the MRI NIMH Macaque template space.

CONCLUSIONS

- Kinetic modeling of [^{11}C]MK-6884 data using arterial and reference region methods can quantify M4 mAChR availability in the brain. SRTM and SRTM2 methods utilizing cerebellum as a reference region are optimally suited for BP_{ND} and RO quantification.
- CVL-231 demonstrates a dose-dependent M4 mAChR receptor occupancy in the striatum of the NHP brain and shows promise for the further development in clinical trials.

Contact information: Vasily Belov, PhD (vbelov@mgh.harvard.edu), Nicolas Guehl, PhD (nguehl@mgh.harvard.edu), Marc Normandin, PhD (normandin@mgh.harvard.edu)